2849

Analysis of the cancer gene targeting of clinical kinase inhibitor drugs by combining cellular and biochemical profiling

Joost C.M. Uitdehaag[1], Jeroen A.D.M. de Roos[1], Antoon M. van Doornmalen[1], Martine B.W. Prinsen[1], Jos de Man[1], Yoshinori Tanizawa[2], Yusuke Kawase[2], Kohichiro Yoshino[2], Rogier C. Buijsman[1], Guido J.R. Zaman[1]

[1]Netherlands Translational Research Center B.V., Molenstraat 110, 5342 CC Oss, The Netherlands; [2]Carna Biosciences Inc., 3rd Floor, BMA, 1-5-5 Minatojima-Minamimachi, Kobe, Japan.

T: +31412700500 **W:** www.ntrc.nl and www.carnabio.com **E:** info@ntrc.nl

Introduction

- We have systematically compared the biochemical and cellular potency and selectivity of all kinase inhibitor drugs on the market
- All kinase inhibitor drugs were profiled in a panel of more than 300 kinase activity assays at Carna Biosciences[1]
- In addition, all kinase inhibitor drugs were profiled in a panel of highly reproducible proliferation assays on 44 well-characterized cell lines (Oncolines™) at NTRC[2]
- The results allow comparison of existing therapies, and define hypotheses for specific target populations for new therapies

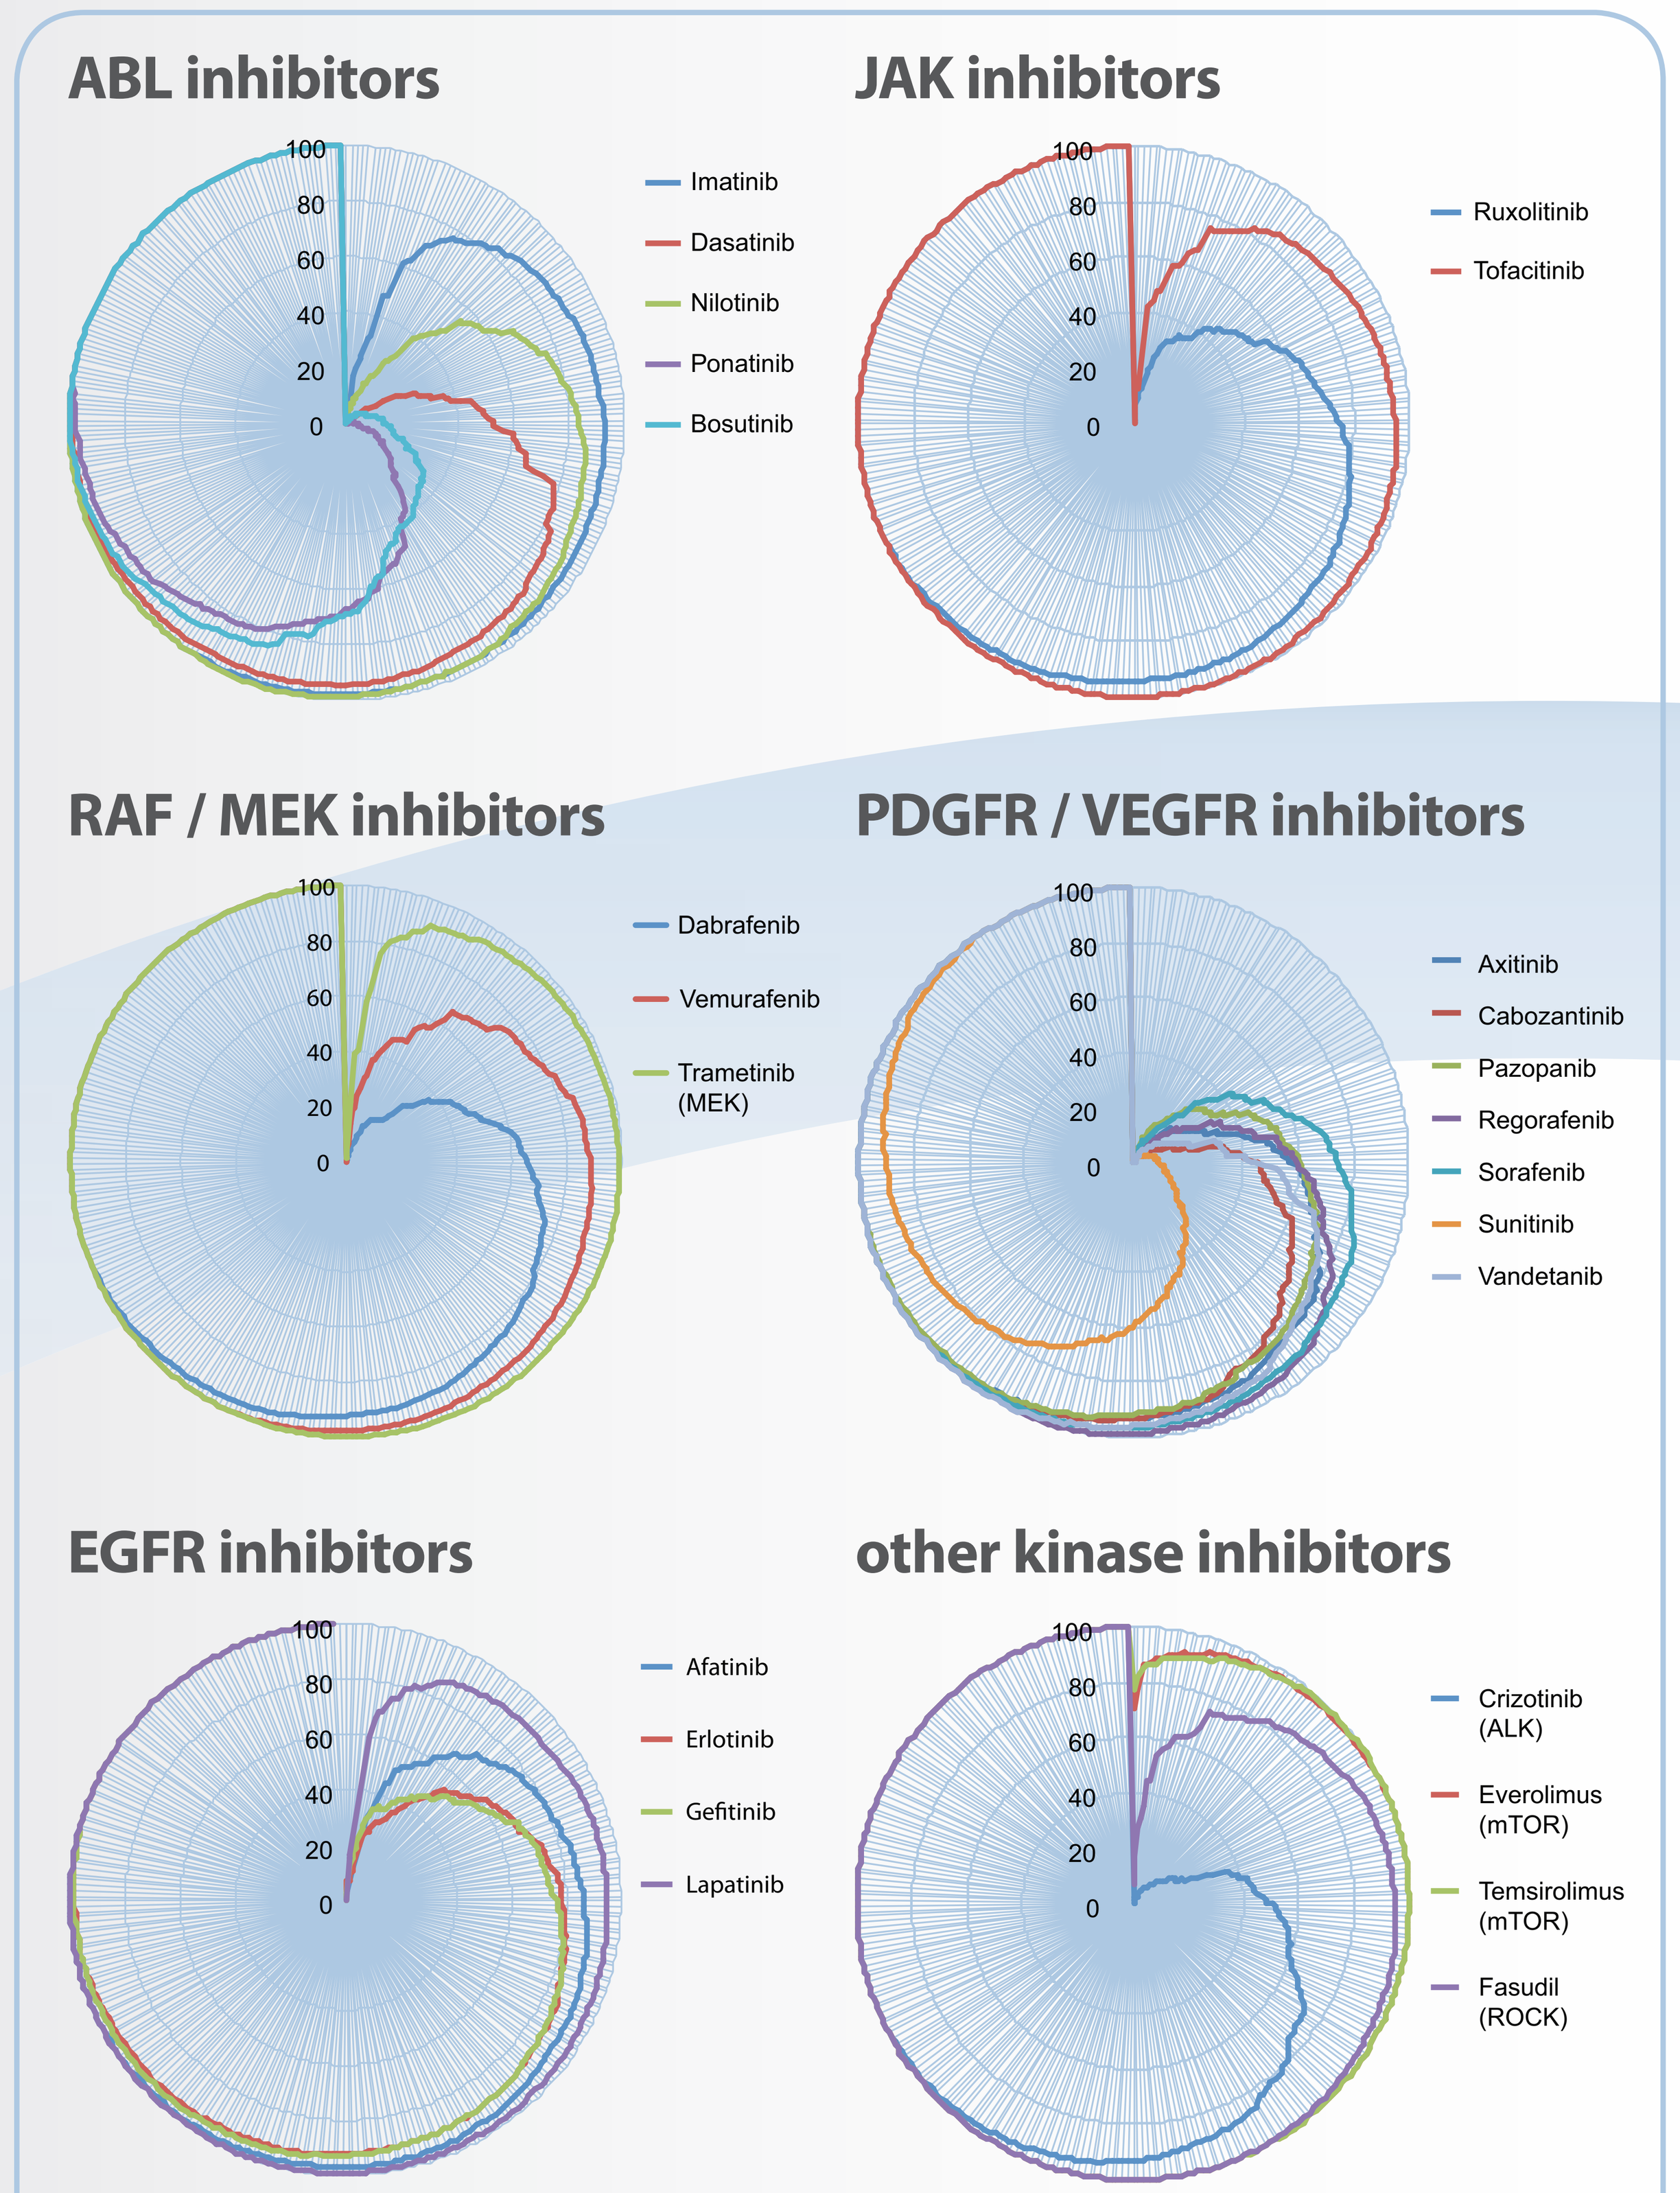
ABL inhibitors
Imatinib
Dasatinib
Nilotinib
Ponatinib
Bosutinib
JAK inhibitors
Ruxolitinib
Tofacitinib
RAF / MEK inhibitors
Dabrafenib
Vemurafenib
Trametinib (MEK)
PDGFR / VEGFR inhibitors
Axitinib
Cabozantinib
Pazopanib
Regorafenib
Sorafenib
Sunitinib
Vandetanib
EGFR inhibitors
Afatinib
Erlotinib
Gefitinib
Lapatinib
other kinase inhibitors
Crizotinib (ALK)
Everolimus (mTOR)
Temsirolimus (mTOR)
Fasudil (ROCK)
0 20 40 60 80 100

Figure 1 *Inhibitors used in this study and their biochemical selectivity profiles.*

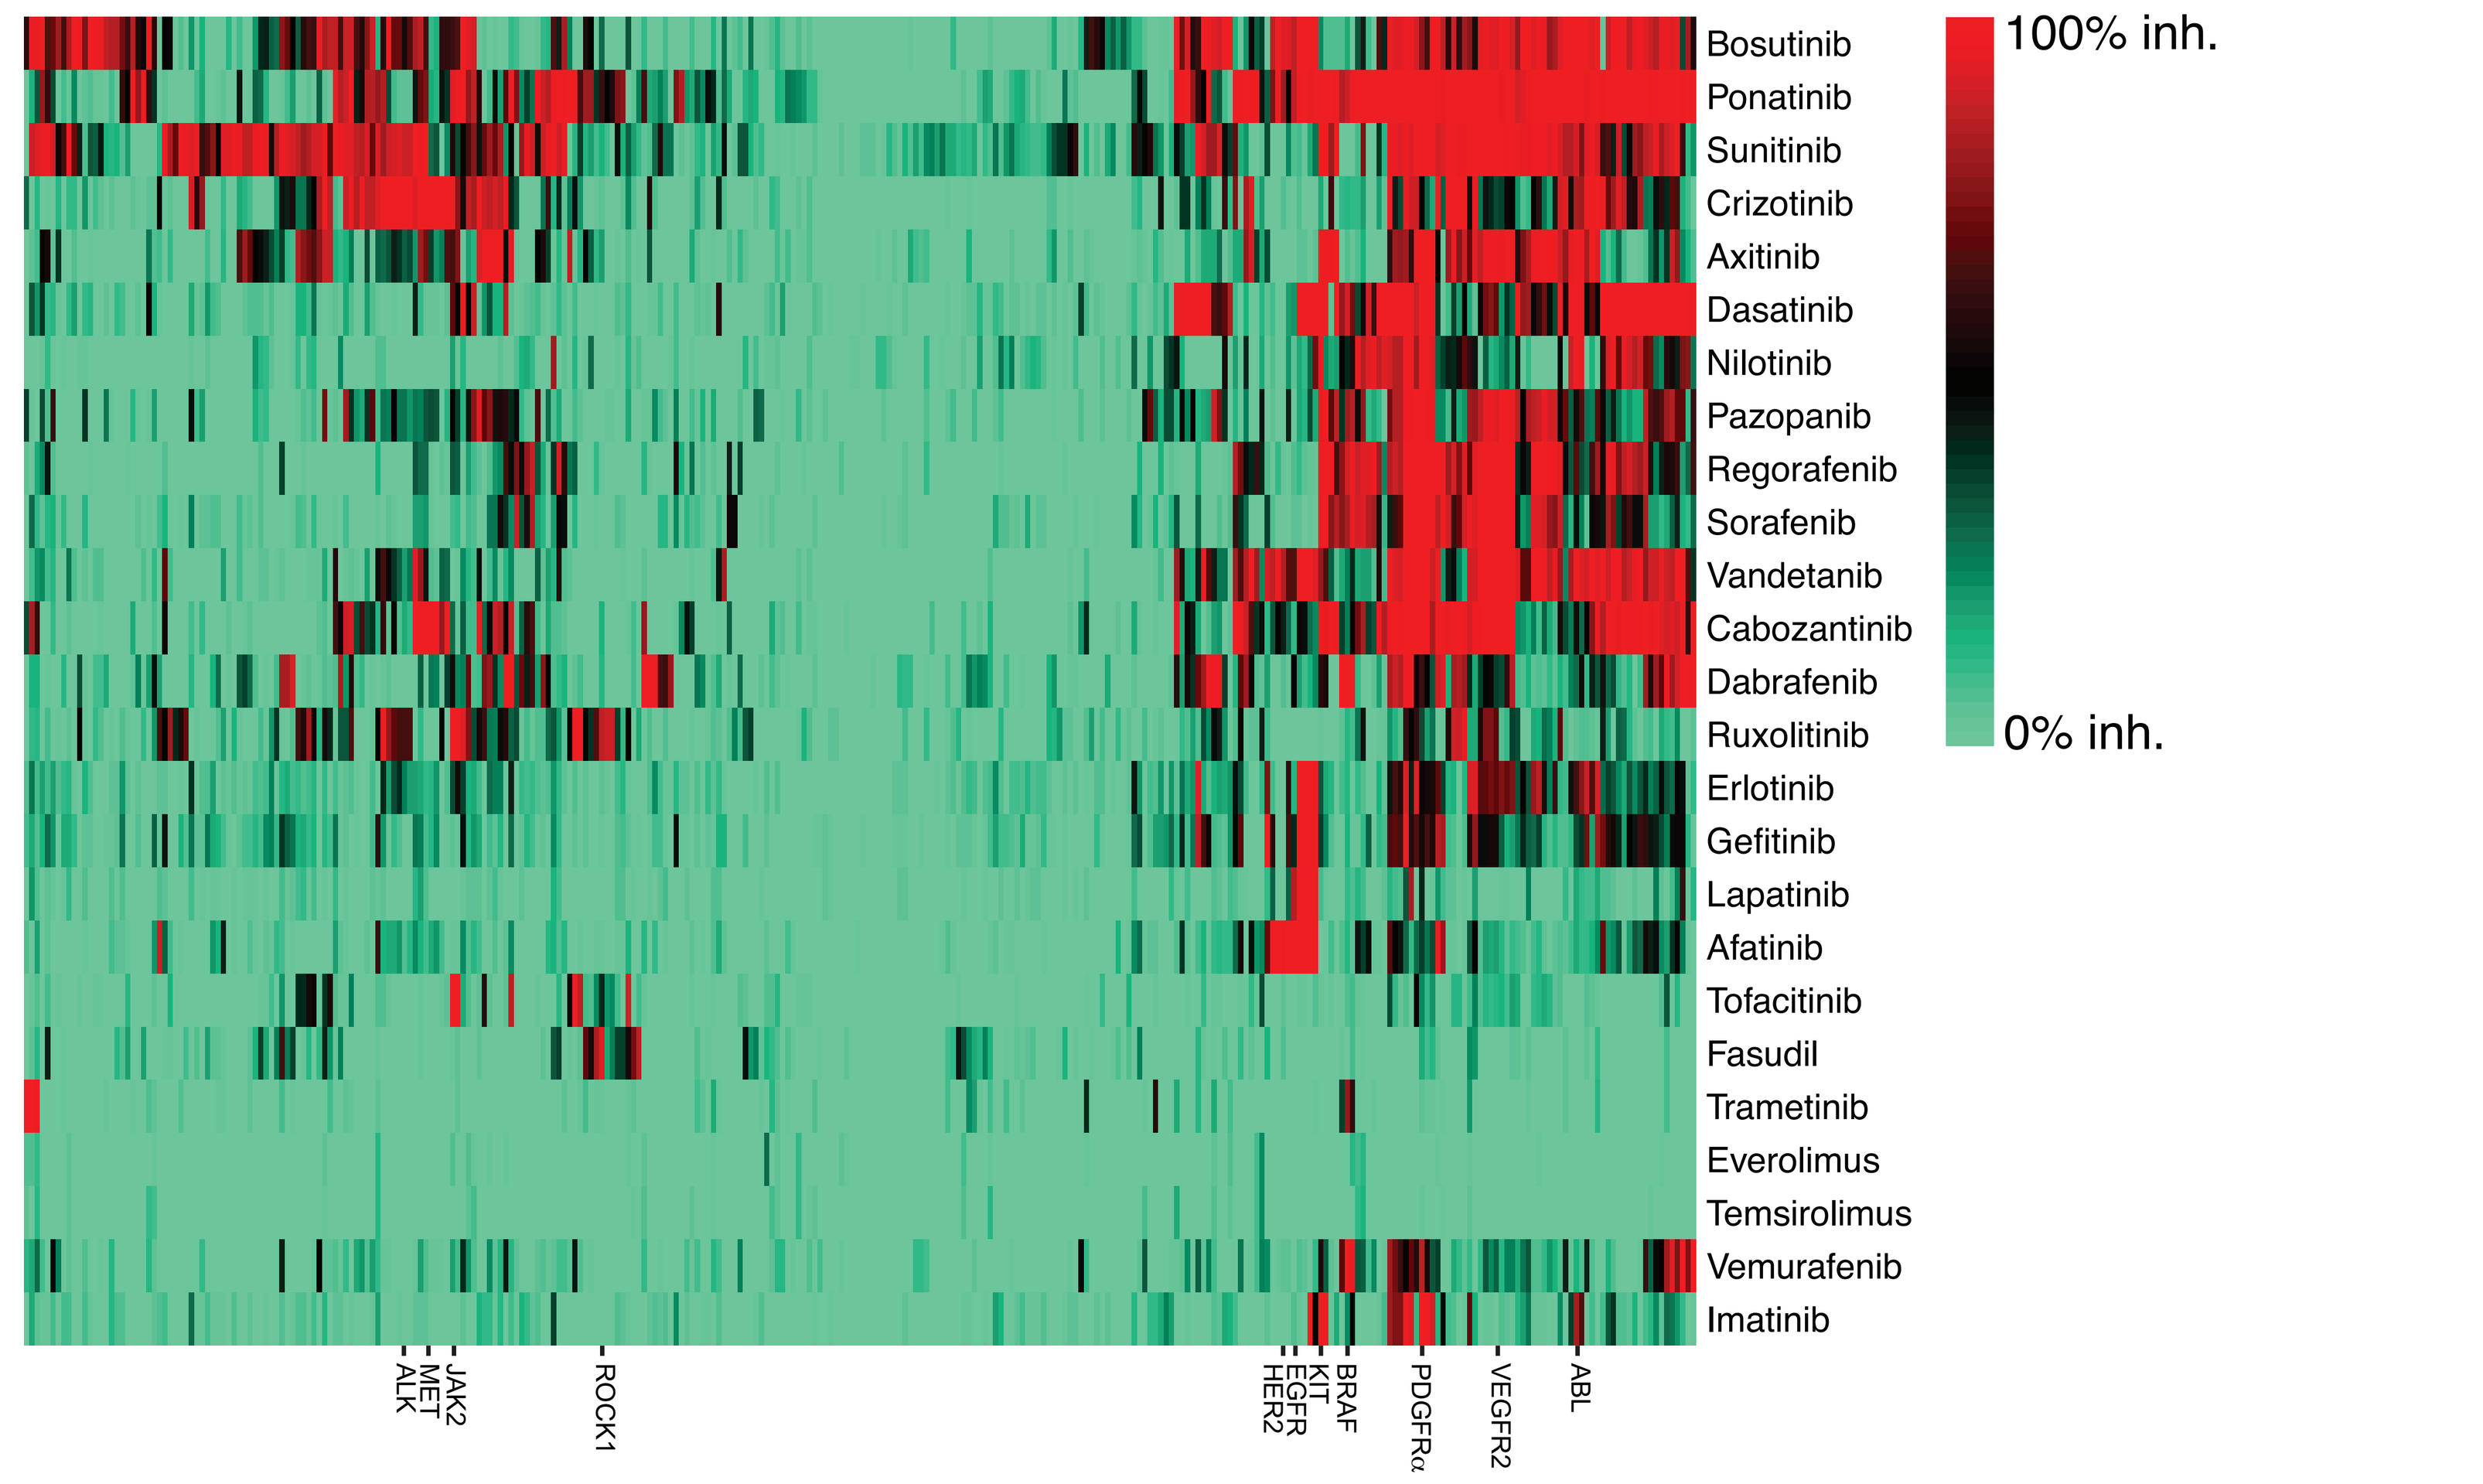
Bosutinib
Ponatinib
Sunitinib
Crizotinib
Axitinib
Dasatinib
Nilotinib
Pazopanib
Regorafenib
Sorafenib
Vandetanib
Cabozantinib
Dabrafenib
Ruxolitinib
Erlotinib
Gefitinib
Lapatinib
Afatinib
Tofacitinib
Fasudil
Trametinib
Everolimus
Temsirolimus
Vemurafenib
Imatinib
100% inh.
0% inh.
ALK MET JAK2 ROCK1 HER2 EGFR KIT BRAF PDGFRα VEGFR2 ABL

Figure 2 *Clustering of the response in kinase activity assays. %-inhibition at 10 µM inhibitor concentration.*

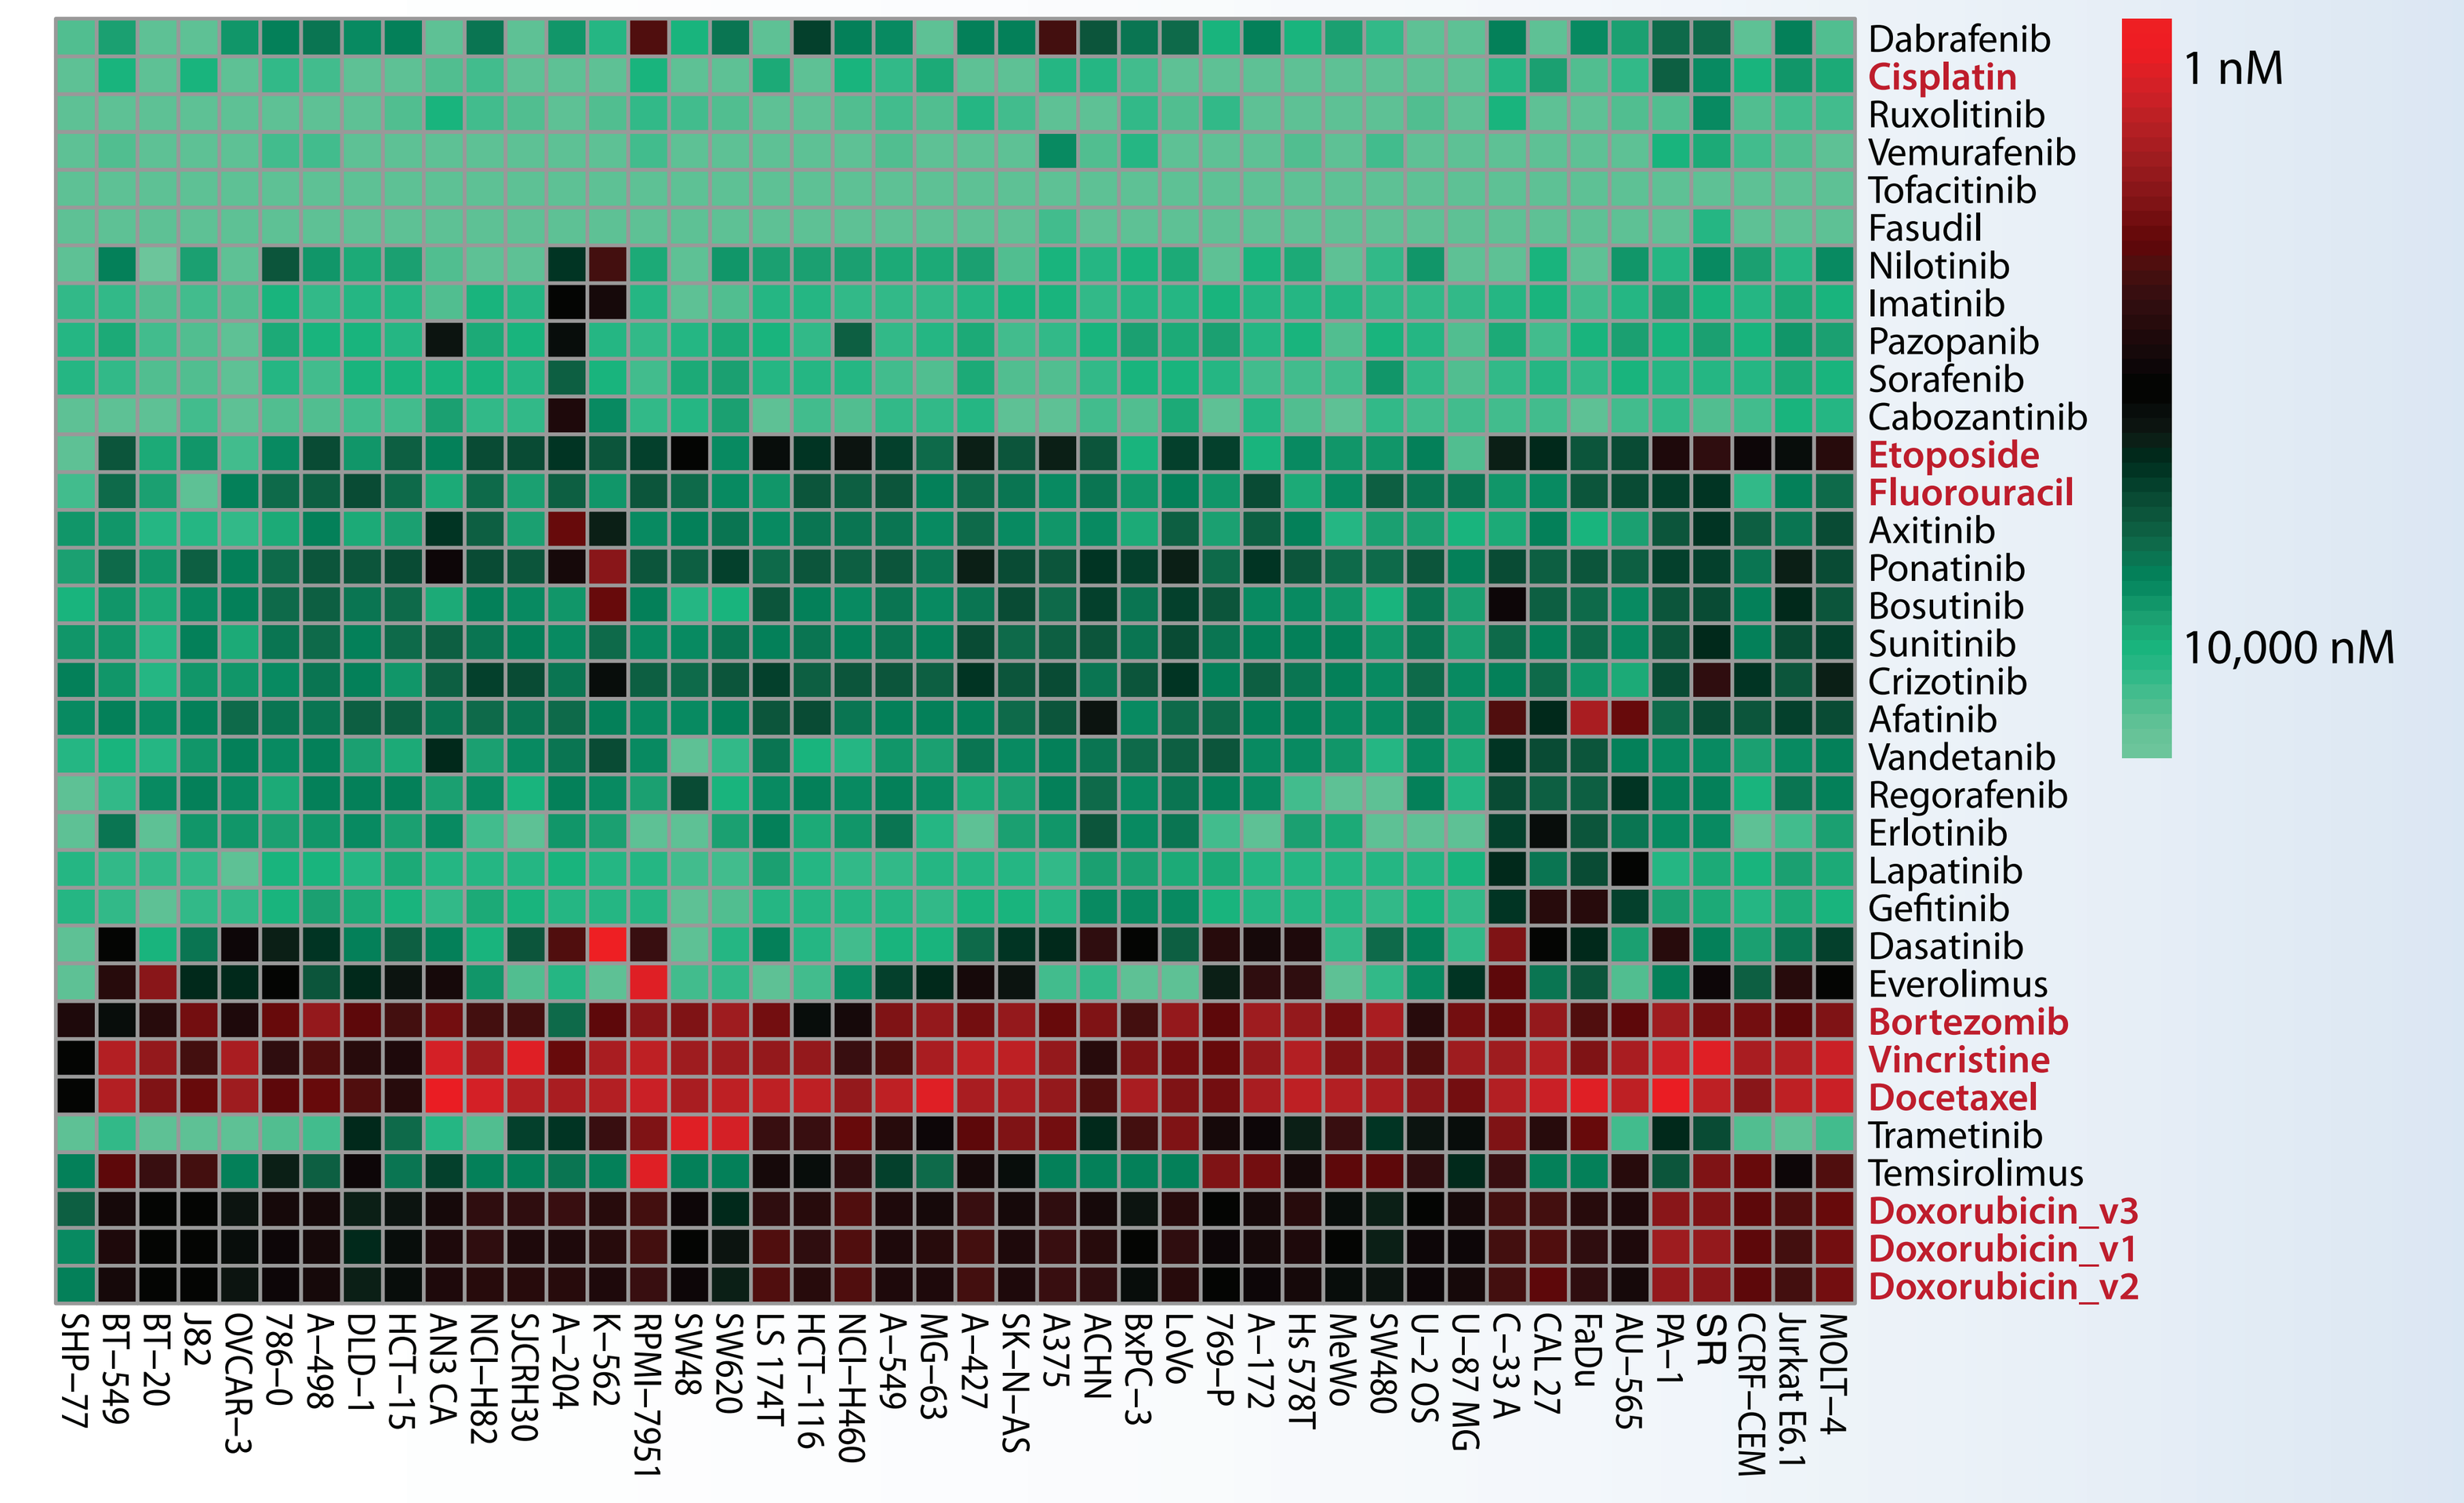
Dabrafenib
Cisplatin
Ruxolitinib
Vemurafenib
Tofacitinib
Fasudil
Nilotinib
Imatinib
Pazopanib
Sorafenib
Cabozantinib
Etoposide
Fluorouracil
Axitinib
Ponatinib
Bosutinib
Sunitinib
Crizotinib
Afatinib
Vandetanib
Regorafenib
Erlotinib
Lapatinib
Gefitinib
Dasatinib
Everolimus
Bortezomib
Vincristine
Docetaxel
Trametinib
Temsirolimus
Doxorubicin_v3
Doxorubicin_v1
Doxorubicin_v2
1 nM
10,000 nM
SHP−77 BT−549 BT−20 J82 OVCAR−3 786−0 A−498 DLD−1 HCT−15 AN3 CA NCI−H82 SJCRH30 A−204 K−562 RPMI−7951 SW48 SW620 LS 174T HCT−116 NCI−H460 A−549 MG−63 A−427 SK−N−AS A375 ACHN BxPC−3 LoVo 769−P A−172 Hs 578T MeWo SW480 U−2 OS U−87 MG C−33 A CAL 27 FaDu AU−565 PA−1 SR CCRF−CEM Jurkat E6.1 MOLT−4

Figure 3 *Clustering of cell proliferation responses ($^{10}logIC_{50}$) In red non kinase drugs.*

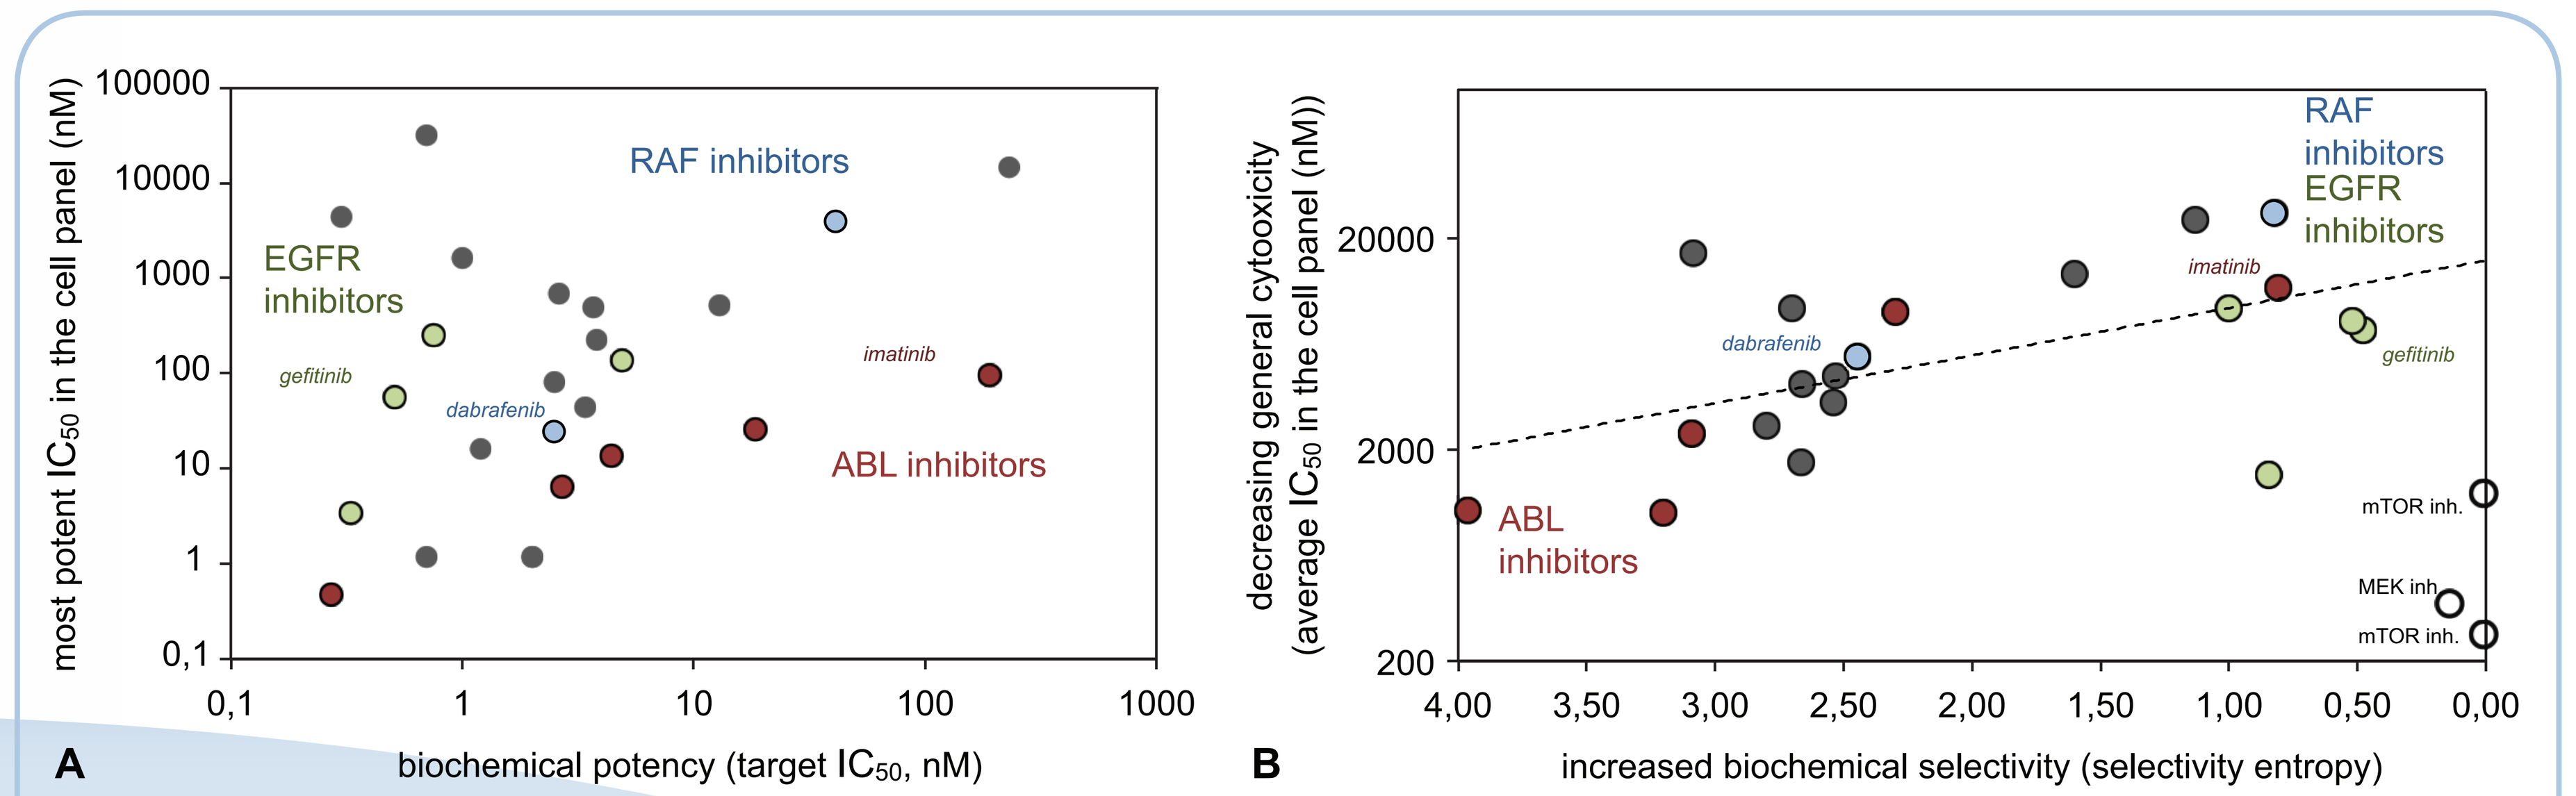
most potent IC50 in the cell panel (nM)
100000 10000 1000 100 10 1 0,1
RAF inhibitors
EGFR inhibitors
gefitinib
dabrafenib
imatinib
ABL inhibitors
0,1 1 10 100 1000
A
biochemical potency (target IC50, nM)
decreasing general cytooxicity (average IC50 in the cell panel (nM))
20000 2000 200
RAF inhibitors
EGFR inhibitors
imatinib
dabrafenib
gefitinib
ABL inhibitors
mTOR inh.
MEK inh.
mTOR inh.
4,00 3,50 3,00 2,50 2,00 1,50 1,00 0,50 0,00
B
increased biochemical selectivity (selectivity entropy)

Figure 4 *Correlation between the biochemical and cellular activities of inhibitors: (**A**) potency (**B**) selectivity.*

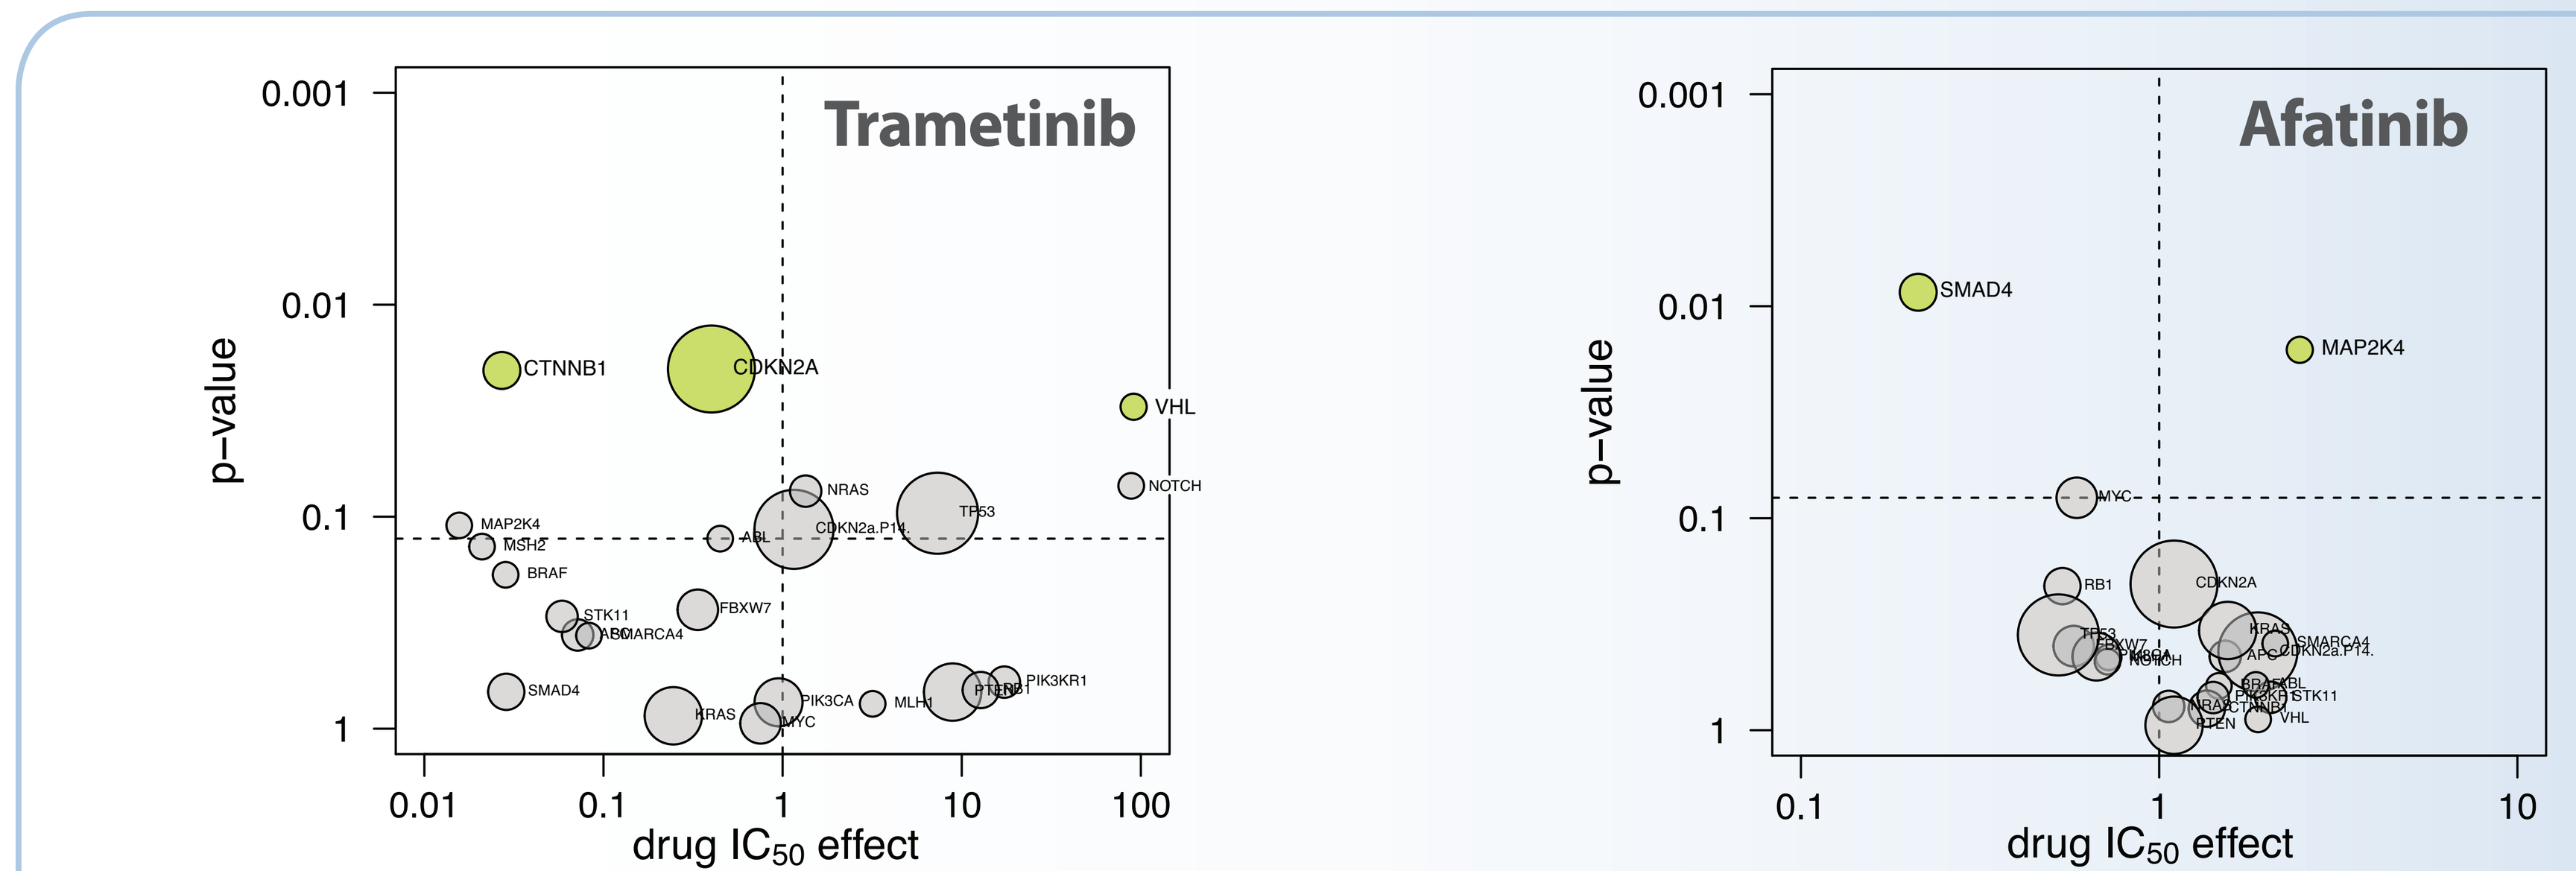
Trametinib
p−value
0.001 0.01 0.1 1
CTNNB1
CDKN2A
VHL
NOTCH
NRAS
TP53
MAP2K4
MSH2
ABL
CDKN2a.P14.
BRAF
STK11
SMARCA4
FBXW7
SMAD4
KRAS
PIK3CA
MYC
MLH1
PIK3KR1
0.01 0.1 1 10 100
drug IC50 effect
Afatinib
SMAD4
MAP2K4
MYC
RB1
CDKN2A
VHL
0.1 1 10
drug IC50 effect

Figure 5 *Anova analysis for drug response markers reveals novel associations for MEK and EGFR inhibitors.*

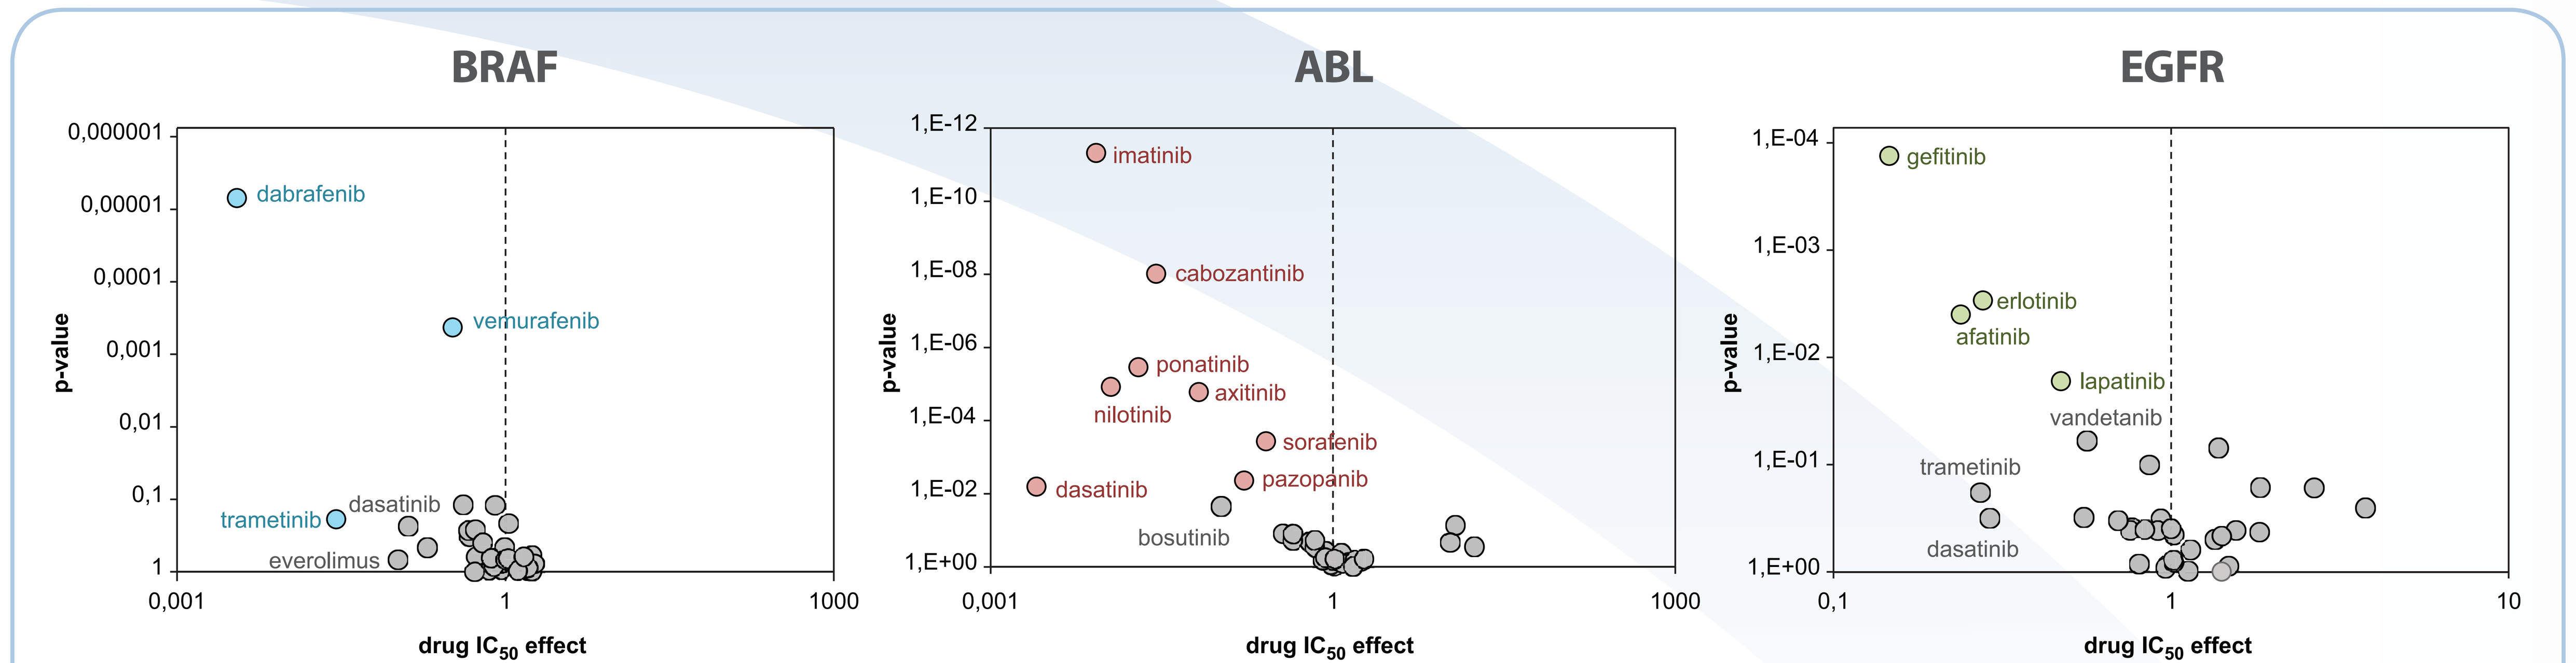
BRAF
p-value
0,000001 0,00001 0,0001 0,001 0,01 0,1 1
dabrafenib
vemurafenib
trametinib
dasatinib
everolimus
0,001 1 1000
drug IC50 effect
ABL
p-value
1,E-12 1,E-10 1,E-08 1,E-06 1,E-04 1,E-02 1,E+00
imatinib
cabozantinib
ponatinib
axitinib
nilotinib
sorafenib
pazopanib
dasatinib
bosutinib
0,001 1 1000
drug IC50 effect
EGFR
p-value
1,E-04 1,E-03 1,E-02 1,E-01 1,E+00
gefitinib
erlotinib
afatinib
lapatinib
vandetanib
trametinib
dasatinib
0,1 1 10
drug IC50 effect

Figure 6 *Comparison of targeting efficacy of marketed BRAF, ABL and EGFR inhibitors.*

References

1. Kitagawa *et al.* Activity-based kinase profiling of approved tyrosine kinase inhibitors. *Genes to cells* **18**: 110-122 (2013).
2. Uitdehaag *et al.* Comparison of the cancer gene targeting and biochemical selectivities of all targeted kinase inhibitors approved for clinical use. *PLOS ONE* **9**: e92146 (2014).

Conclusions

- Cell line panel profiling reveals that MEK inhibitors can be beneficial in β-catenin mutant cancers and EGFR inhibitors in SMAD4 mutant cancers
- Biochemical selectivity and potency are prime determinants of the target efficacy of kinase inhibitor drugs
- Integrated biochemical and cellular profiling can help to prioritize compounds and provide directions for optimization

NETHERLANDS TRANSLATIONAL RESEARCH CENTER B.V.